Enhancing the Cleo experiment simulation testbed to support all-optical control, multi-channel optogenetics, and easier integration into data analysis pipelines

Kyle A. Johnsen[1,2] (kjohnsen@gatech.edu), Nathanael A. Cruzado[1], Adam S. Charles[3], Christopher J. Rozell[1]

1. Georgia Institute of Technology, Atlanta, GA, USA 2. Emory University, Atlanta, GA, USA 3. The Johns Hopkins University, Baltimore, MD, USA

Simulate complex ephys, optogenetics, and 2P imaging experiments with ease

Introduction

Cleo turns a spiking neural network model into a virtual experiment—useful for experiment design and model evaluation:

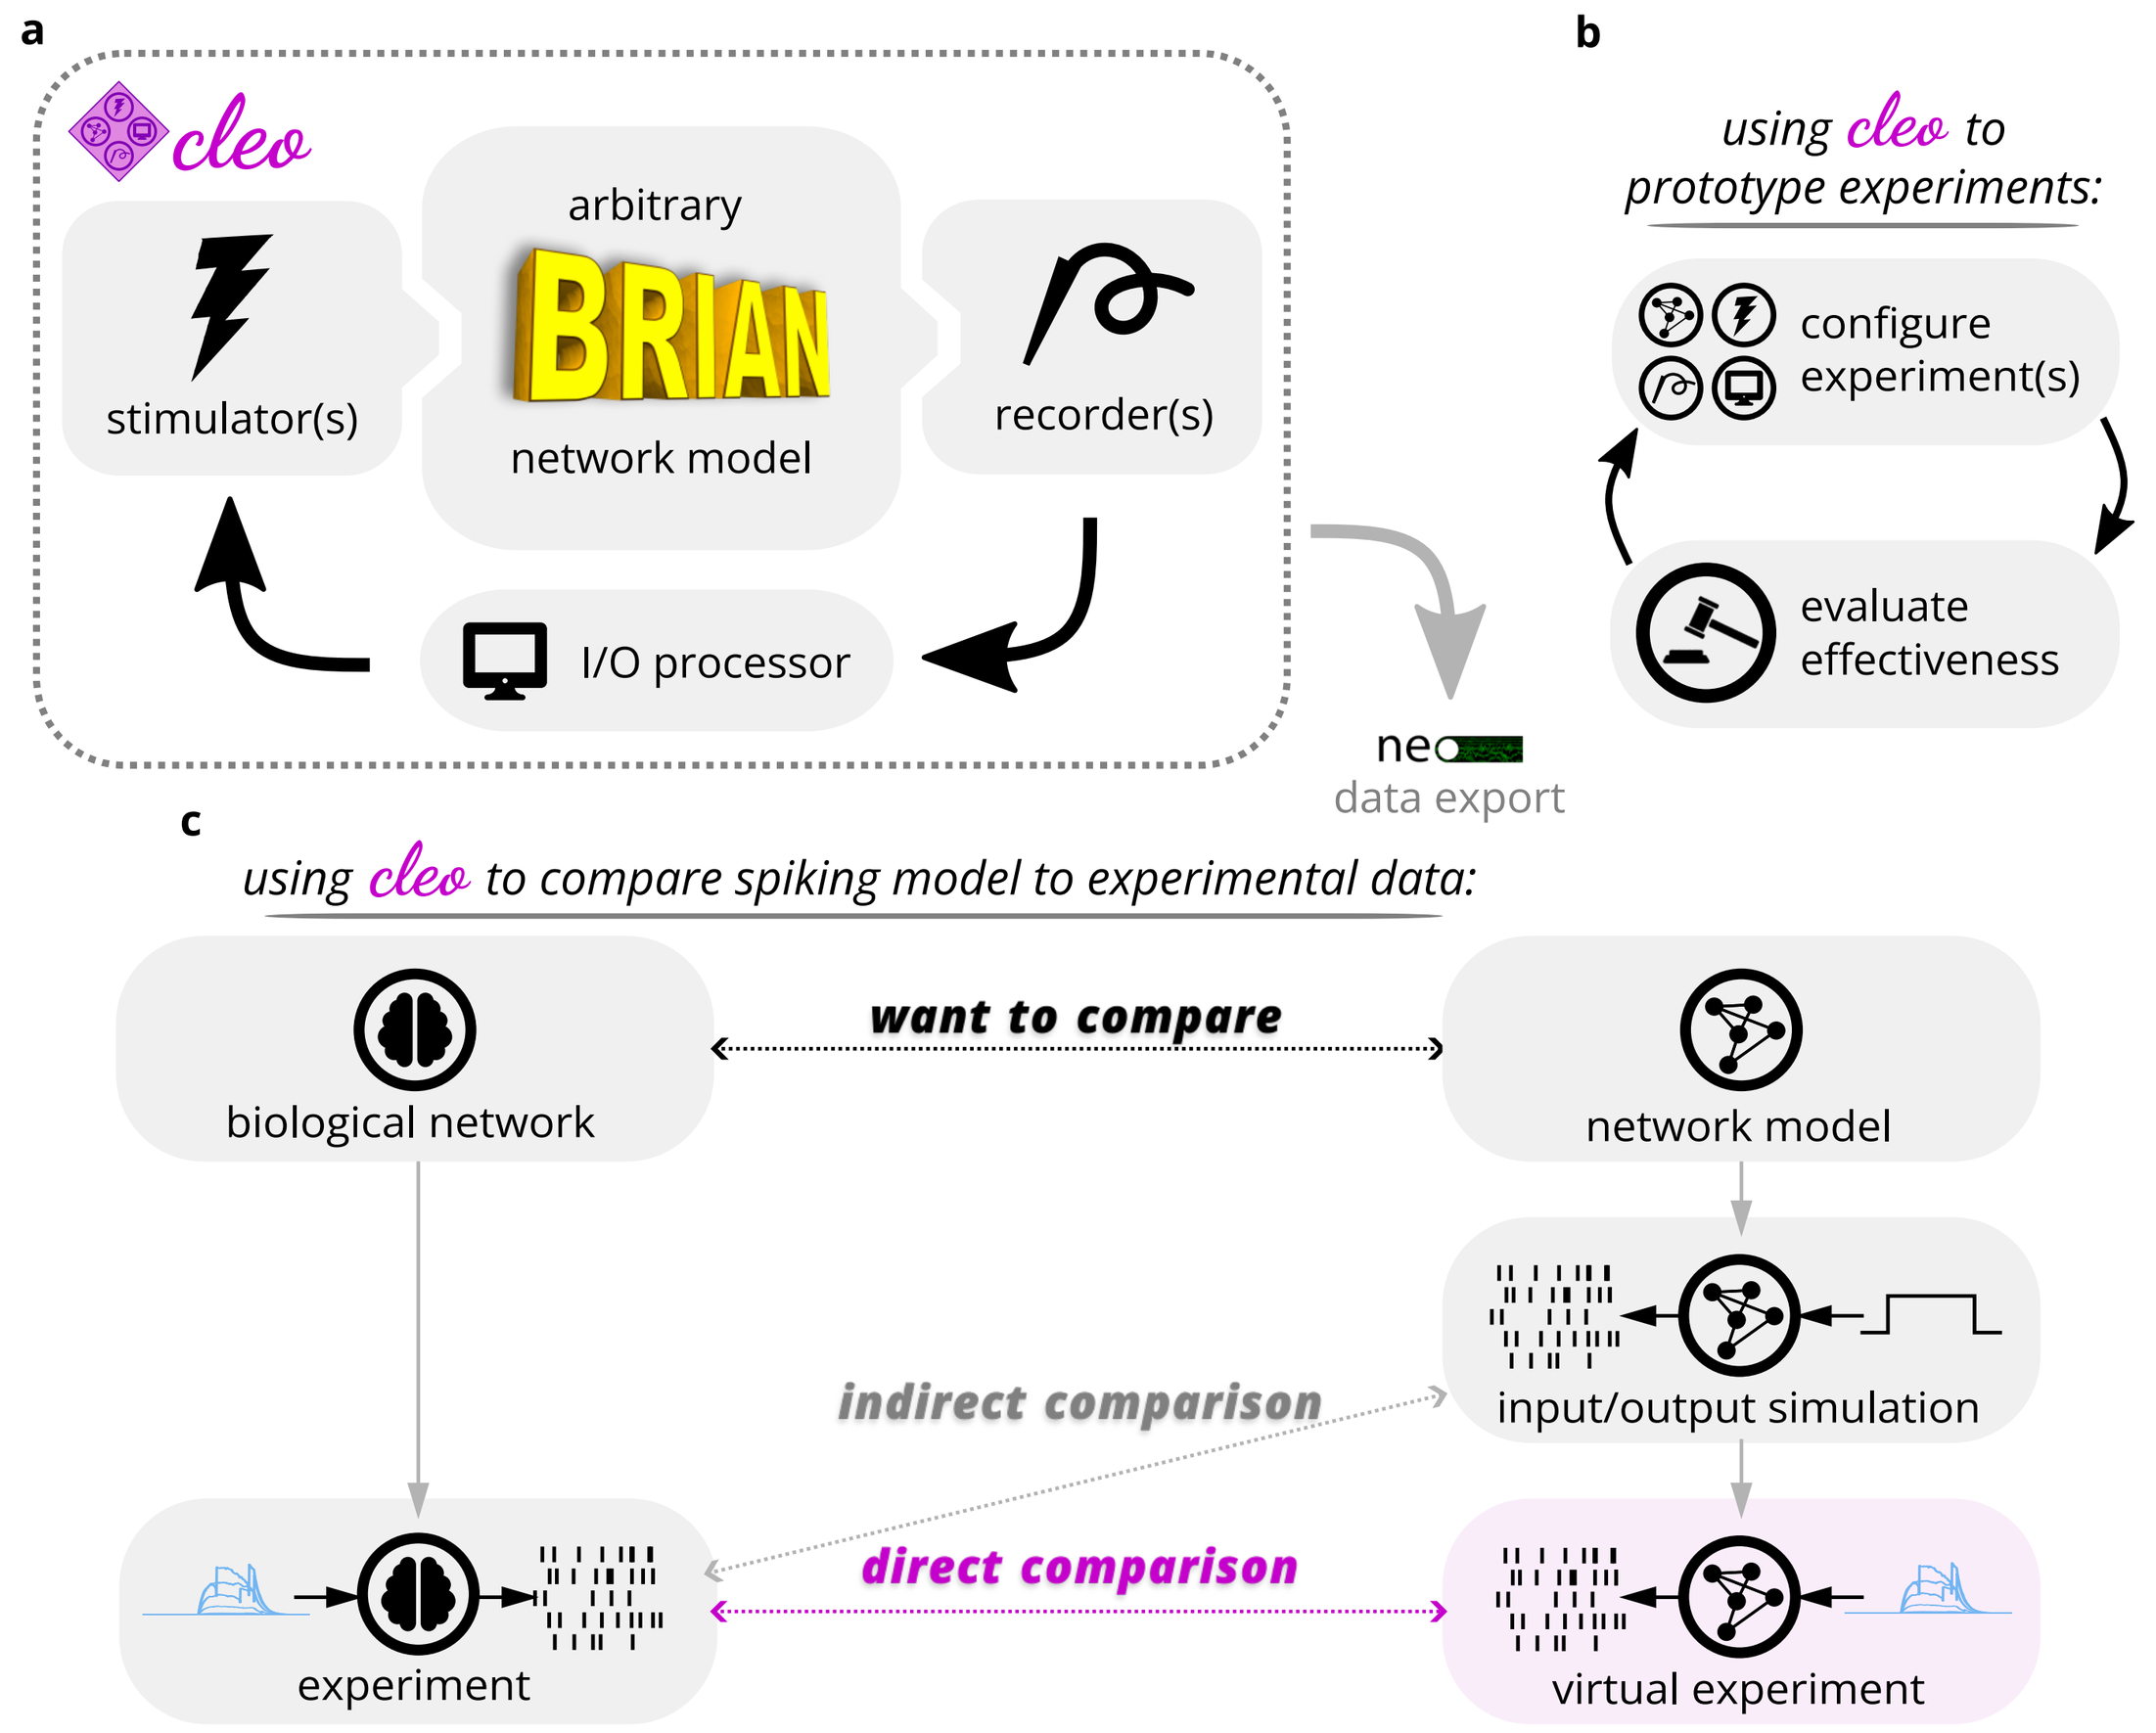
a
cleo
arbitrary
BRIAN
network model
stimulator(s)
recorder(s)
I/O processor
neo
data export
b
using cleo to prototype experiments:
configure experiment(s)
evaluate effectiveness
c
using cleo to compare spiking model to experimental data:
biological network
want to compare
network model
input/output simulation
indirect comparison
direct comparison
experiment
virtual experiment

It does this by simulating various components of the experimental interface (new features in green):

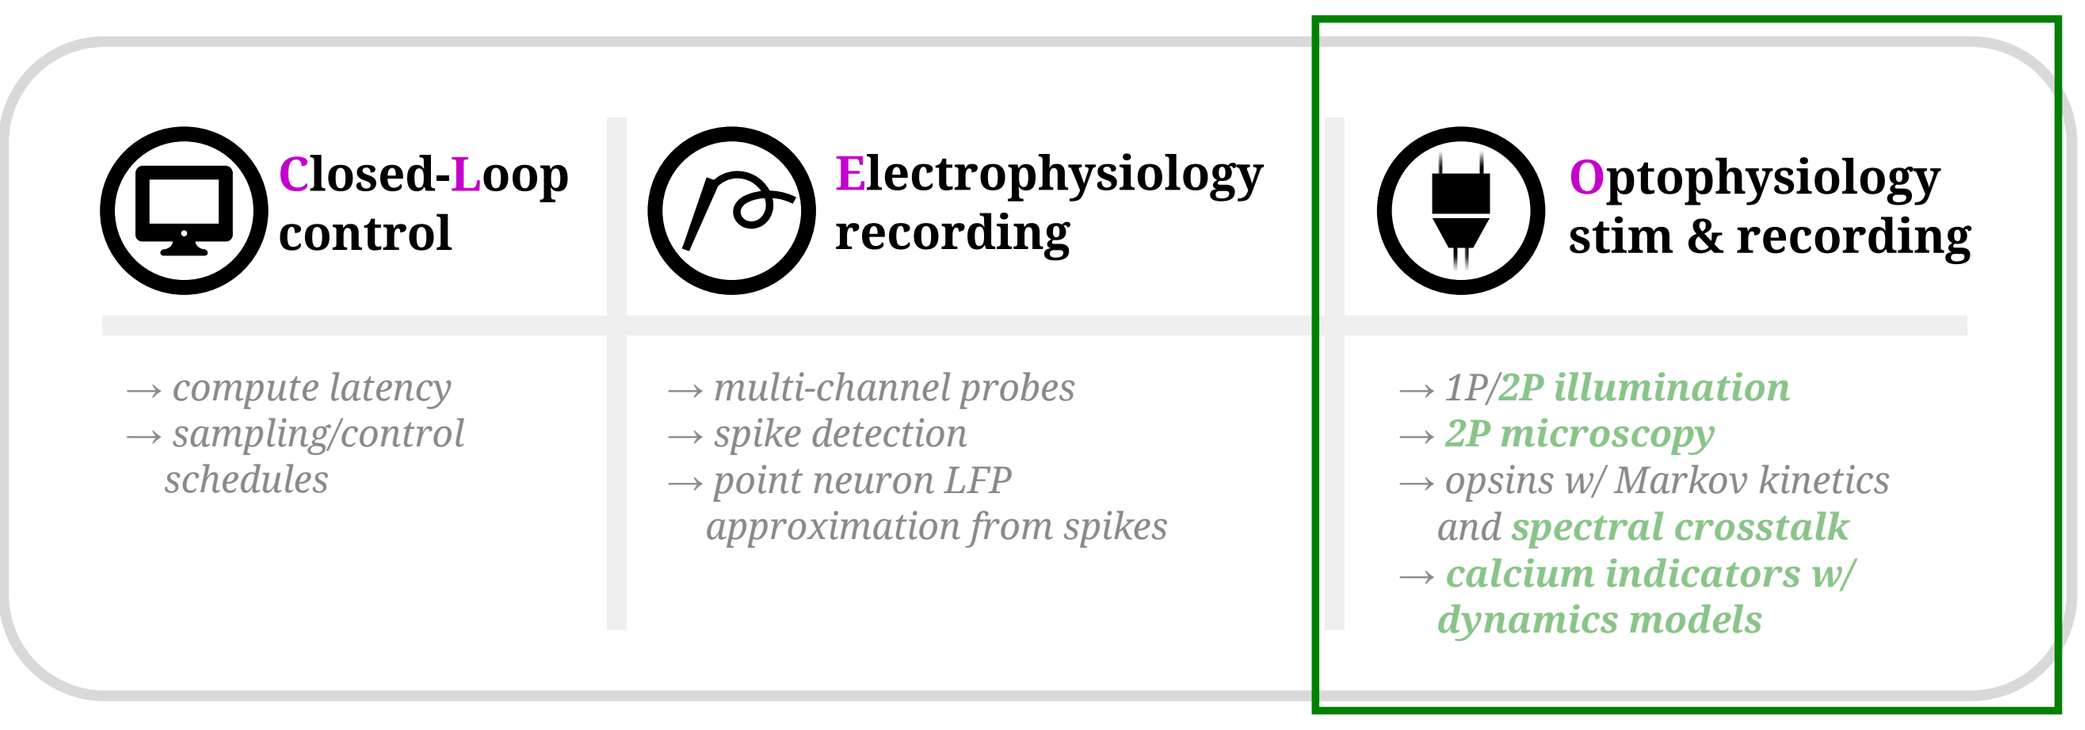
Closed-Loop control
→ compute latency
→ sampling/control schedules
Electrophysiology recording
→ multi-channel probes
→ spike detection
→ point neuron LFP approximation from spikes
Optophysiology stim & recording
→ 1P/2P illumination
→ 2P microscopy
→ opsins w/ Markov kinetics and spectral crosstalk
→ calcium indicators w/ dynamics models

Cleo is part of a larger effort developing and applying closed-loop optogenetic control (CLOC) methods:

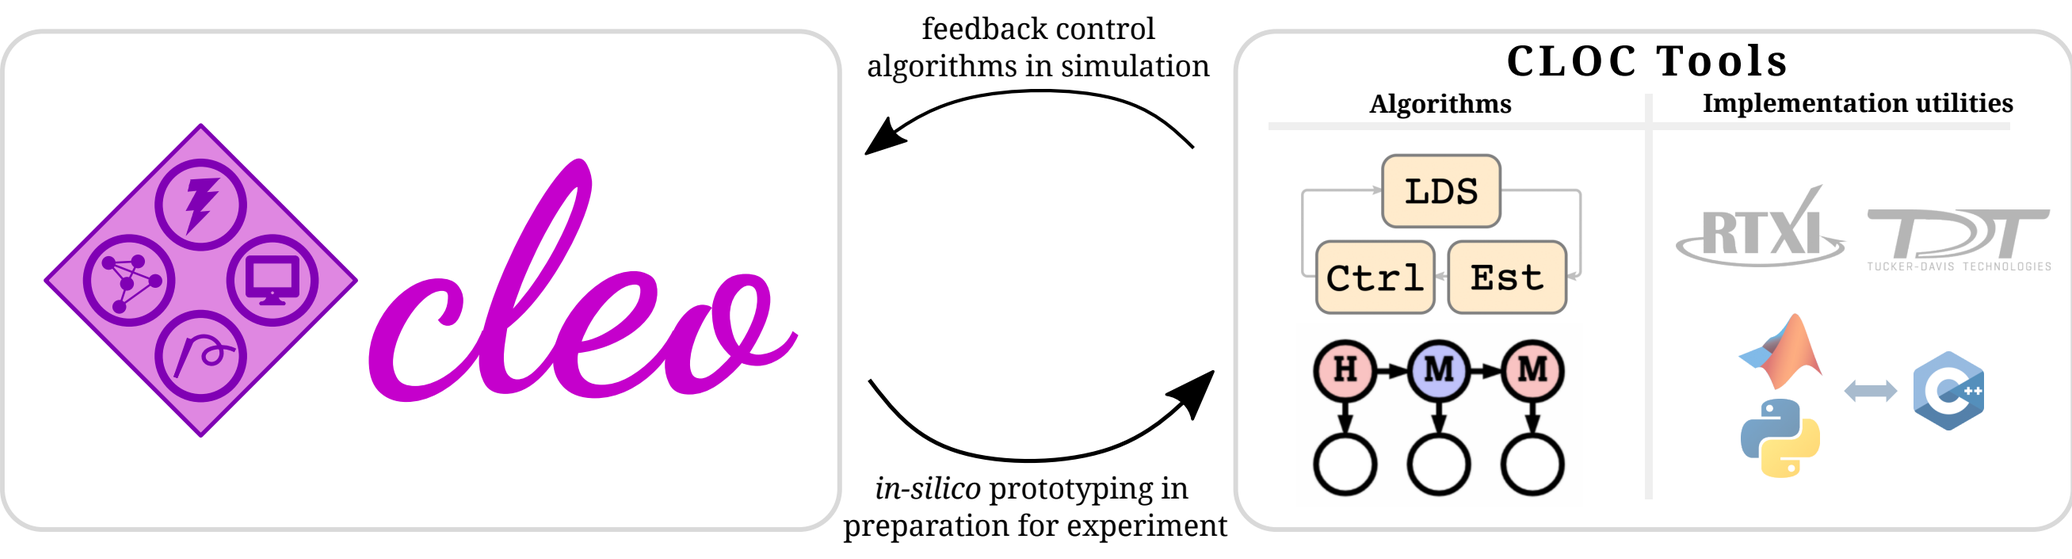
cleo
feedback control algorithms in simulation
in-silico prototyping in preparation for experiment
CLOC Tools
Algorithms
Implementation utilities
LDS
Ctrl
Est

See `https://cloctools.github.io` and Willats et al. [1, 2, 3], Bolus et al. [4, 5].

Electrophysiology

Virtual electrode contacts record spiking and local field potentials [6] from the network:

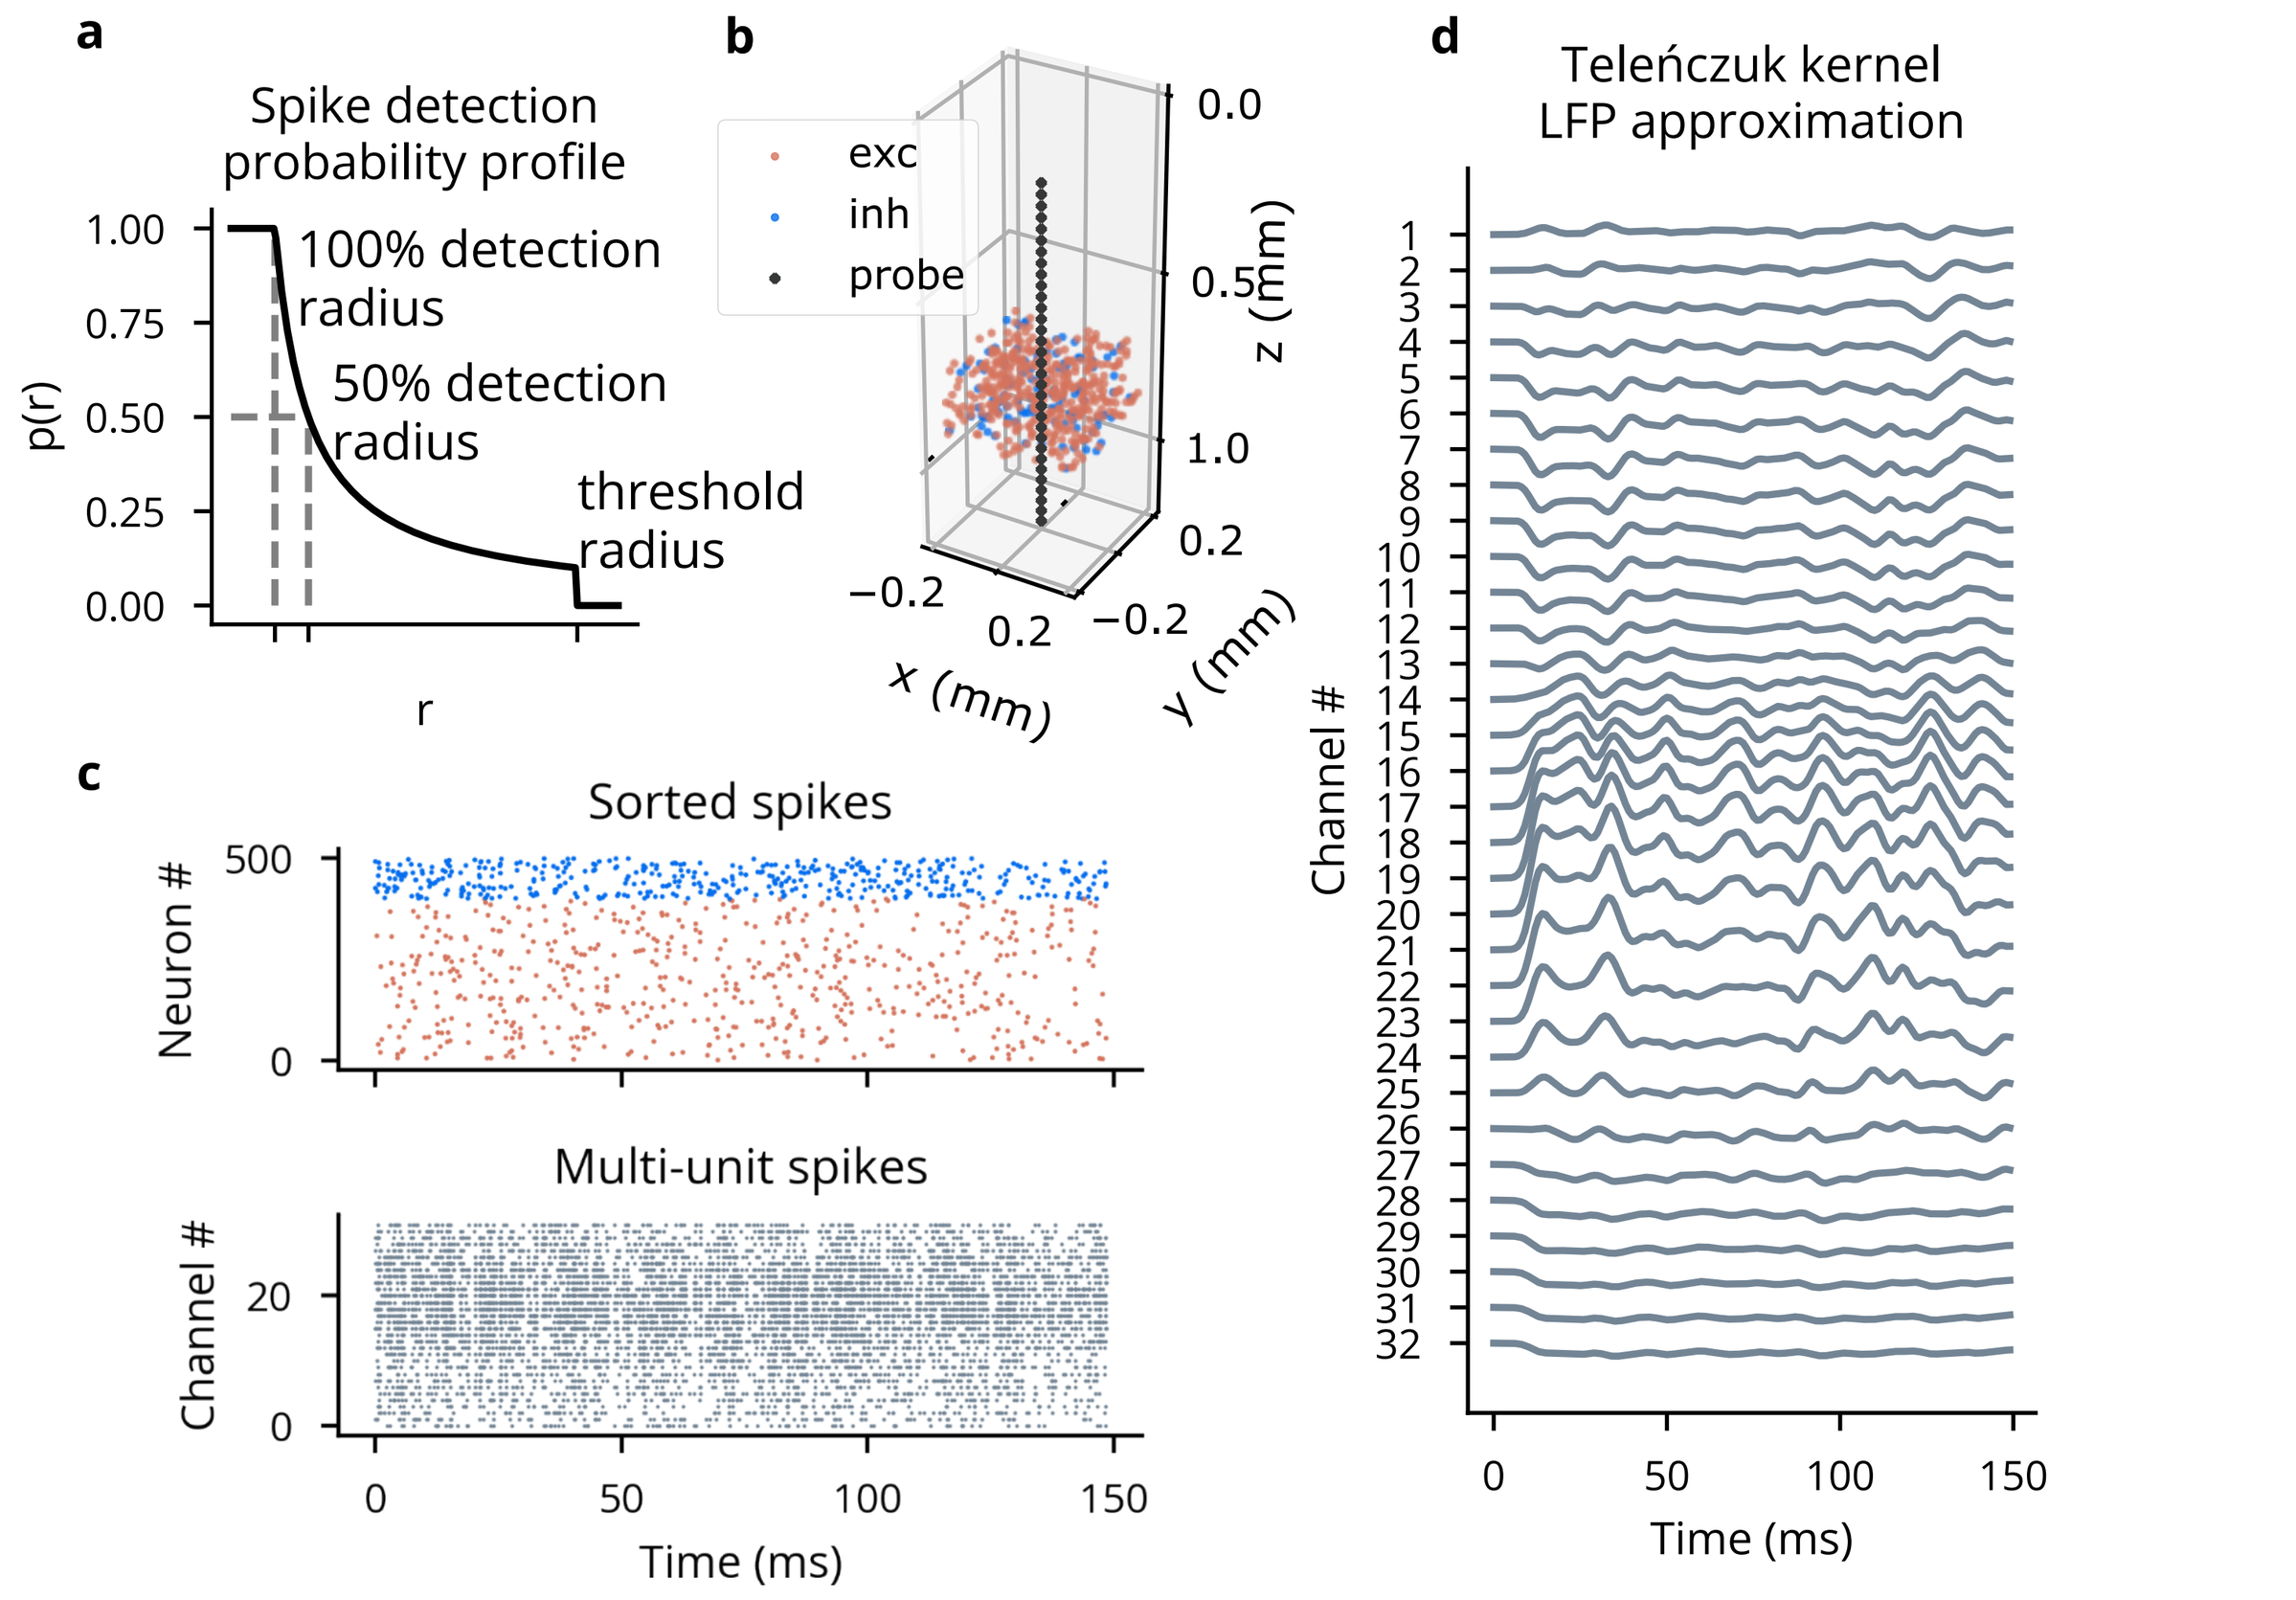
a
Spike detection probability profile
100% detection radius
50% detection radius
threshold radius
p(r)
r
b
exc
inh
probe
x (mm)
y (mm)
z (mm)
c
Sorted spikes
Neuron #
Multi-unit spikes
Channel #
Time (ms)
d
Teleńczuk kernel LFP approximation
Channel #
Time (ms)

Optogenetics

Light propagation and opsin kinetics models enable simulation of one-photon optogenetics [7]:

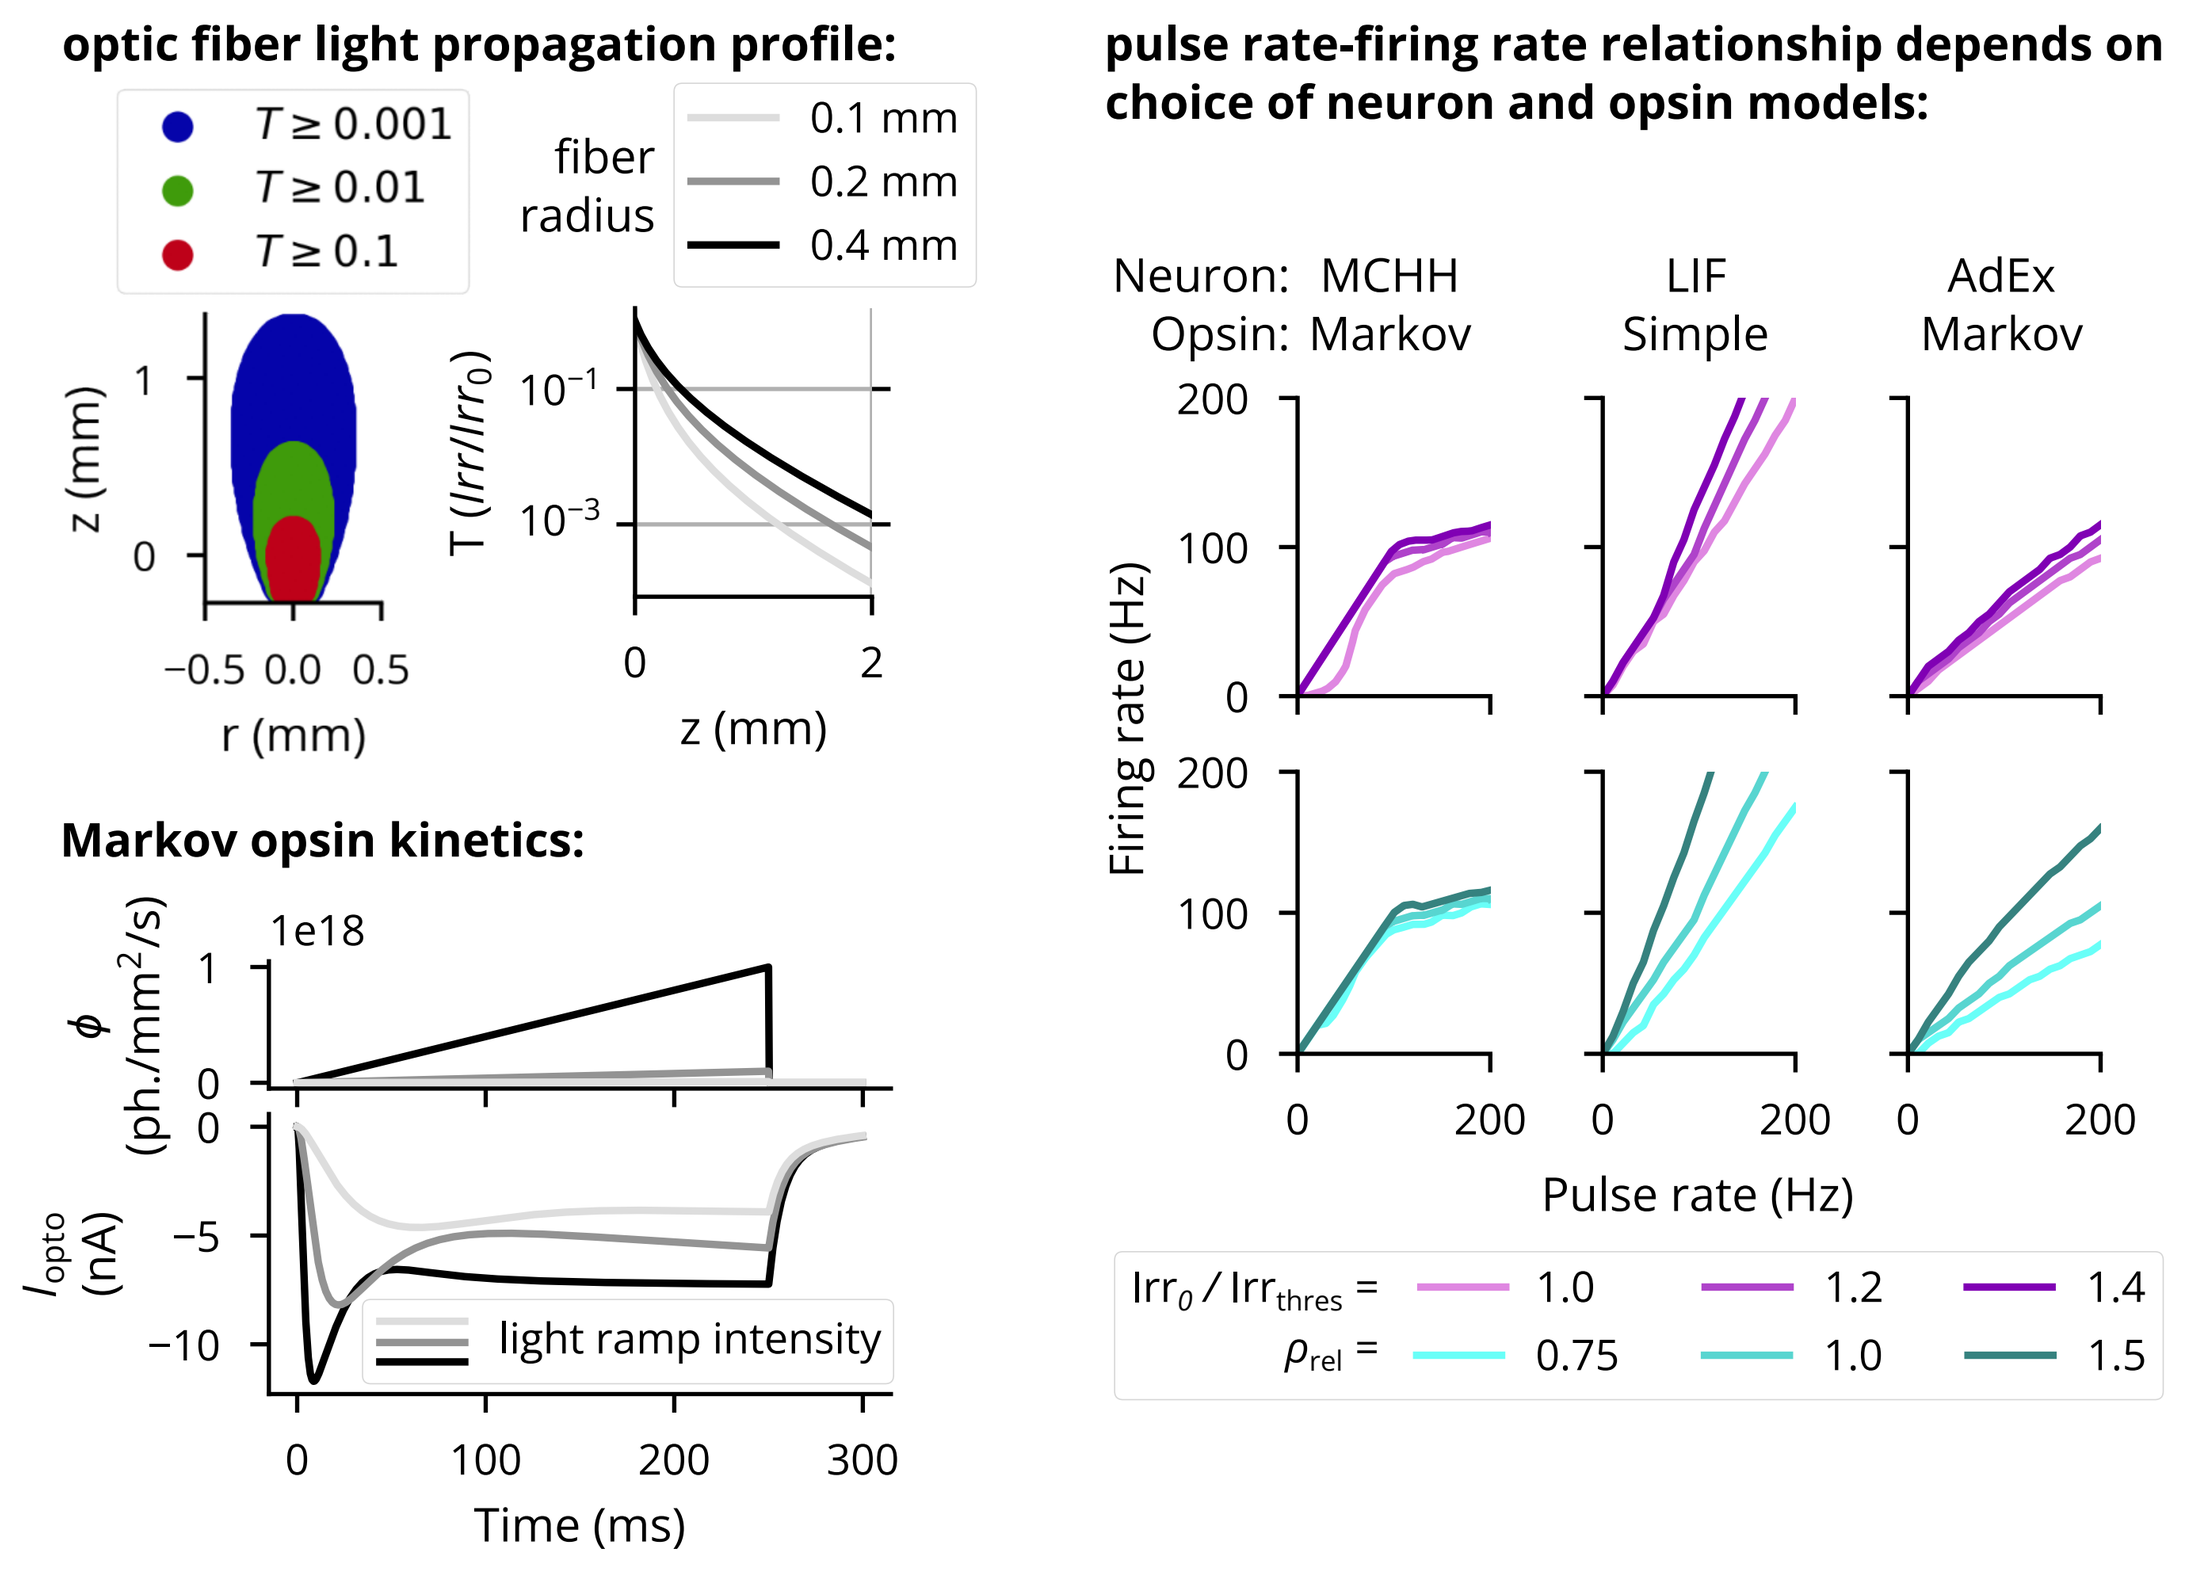
optic fiber light propagation profile:
$T \geq 0.001$
$T \geq 0.01$
$T \geq 0.1$
fiber radius
0.1 mm
0.2 mm
0.4 mm
z (mm)
r (mm)
T (Irr/Irr₀)
Markov opsin kinetics:
light ramp intensity
Time (ms)
pulse rate-firing rate relationship depends on choice of neuron and opsin models:
Neuron: MCHH Opsin: Markov
LIF Simple
AdEx Markov
Firing rate (Hz)
Pulse rate (Hz)

Cleo also simulates multi-site/opsin stimulation, including crosstalk from overlapping action spectra [8]:

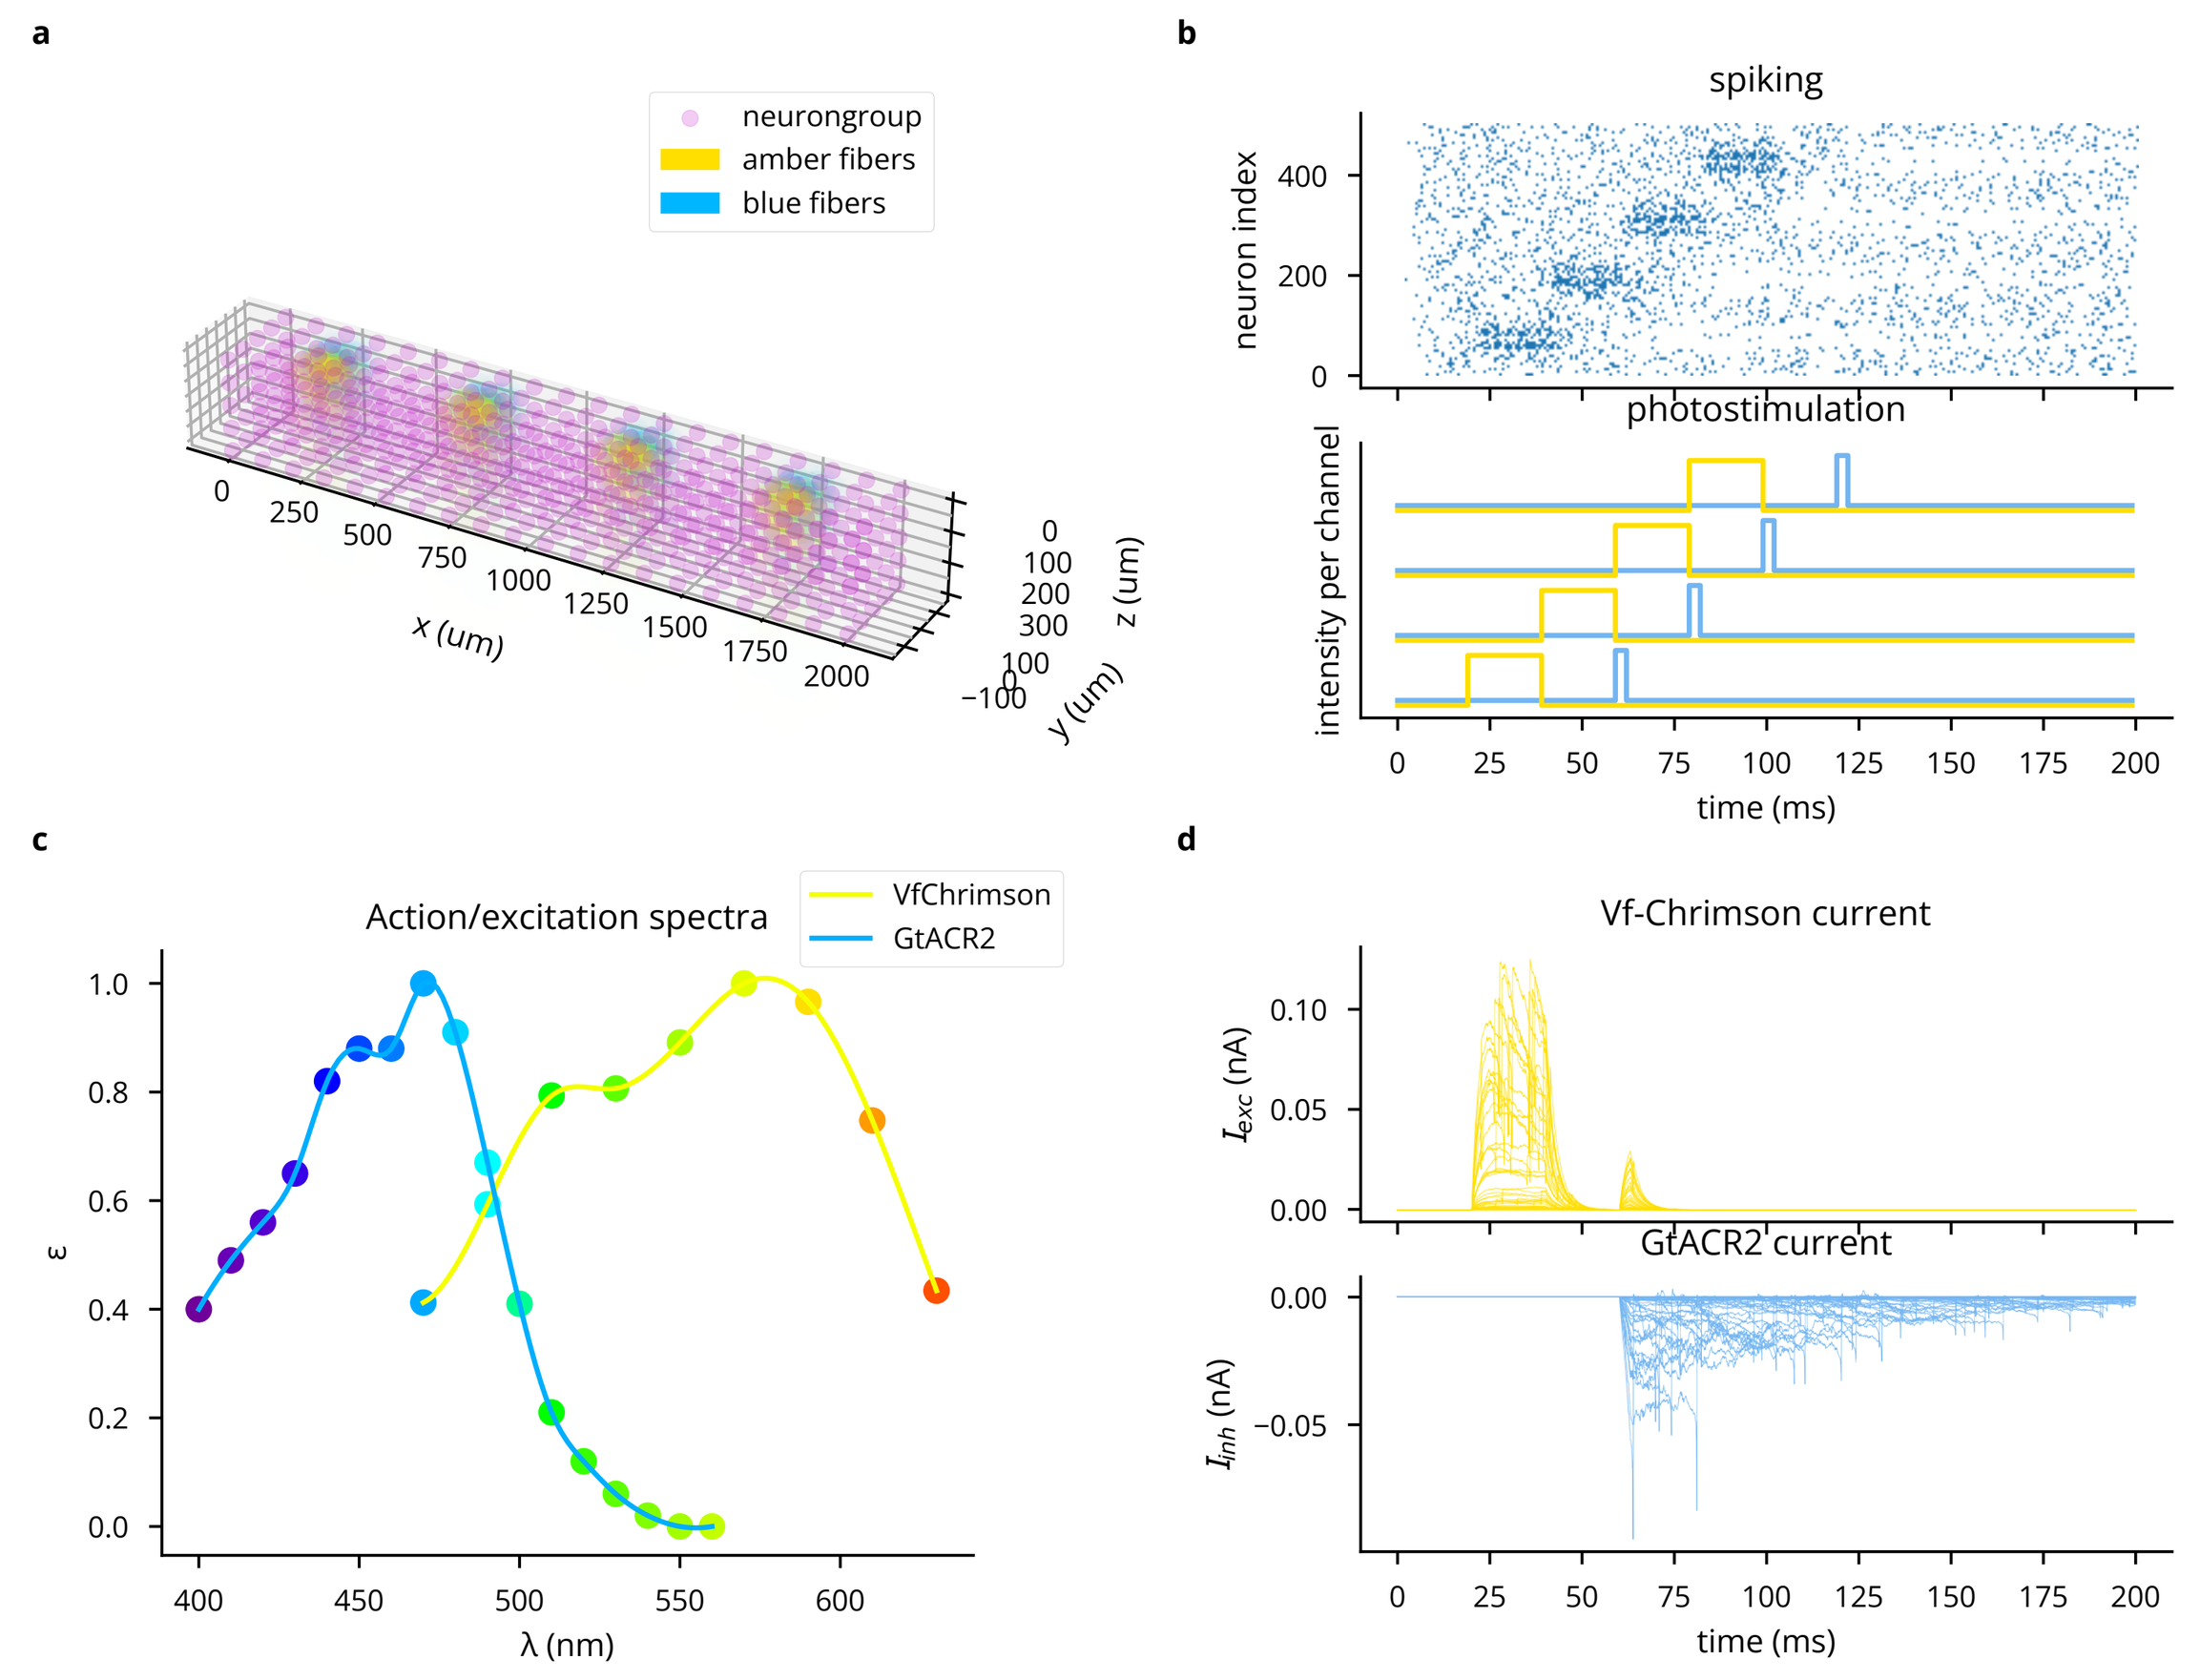
a
neurongroup
amber fibers
blue fibers
x (um)
y (um)
z (um)
b
spiking
neuron index
photostimulation
intensity per channel
time (ms)
c
Action/excitation spectra
VfChrimson
GtACR2
λ (nm)
d
Vf-Chrimson current
GtACR2 current
time (ms)

Two-photon imaging and photostimulation

A microscope selects neurons in focus for imaging while a Gaussian ellipsoid models off-target 2P laser stimulation:

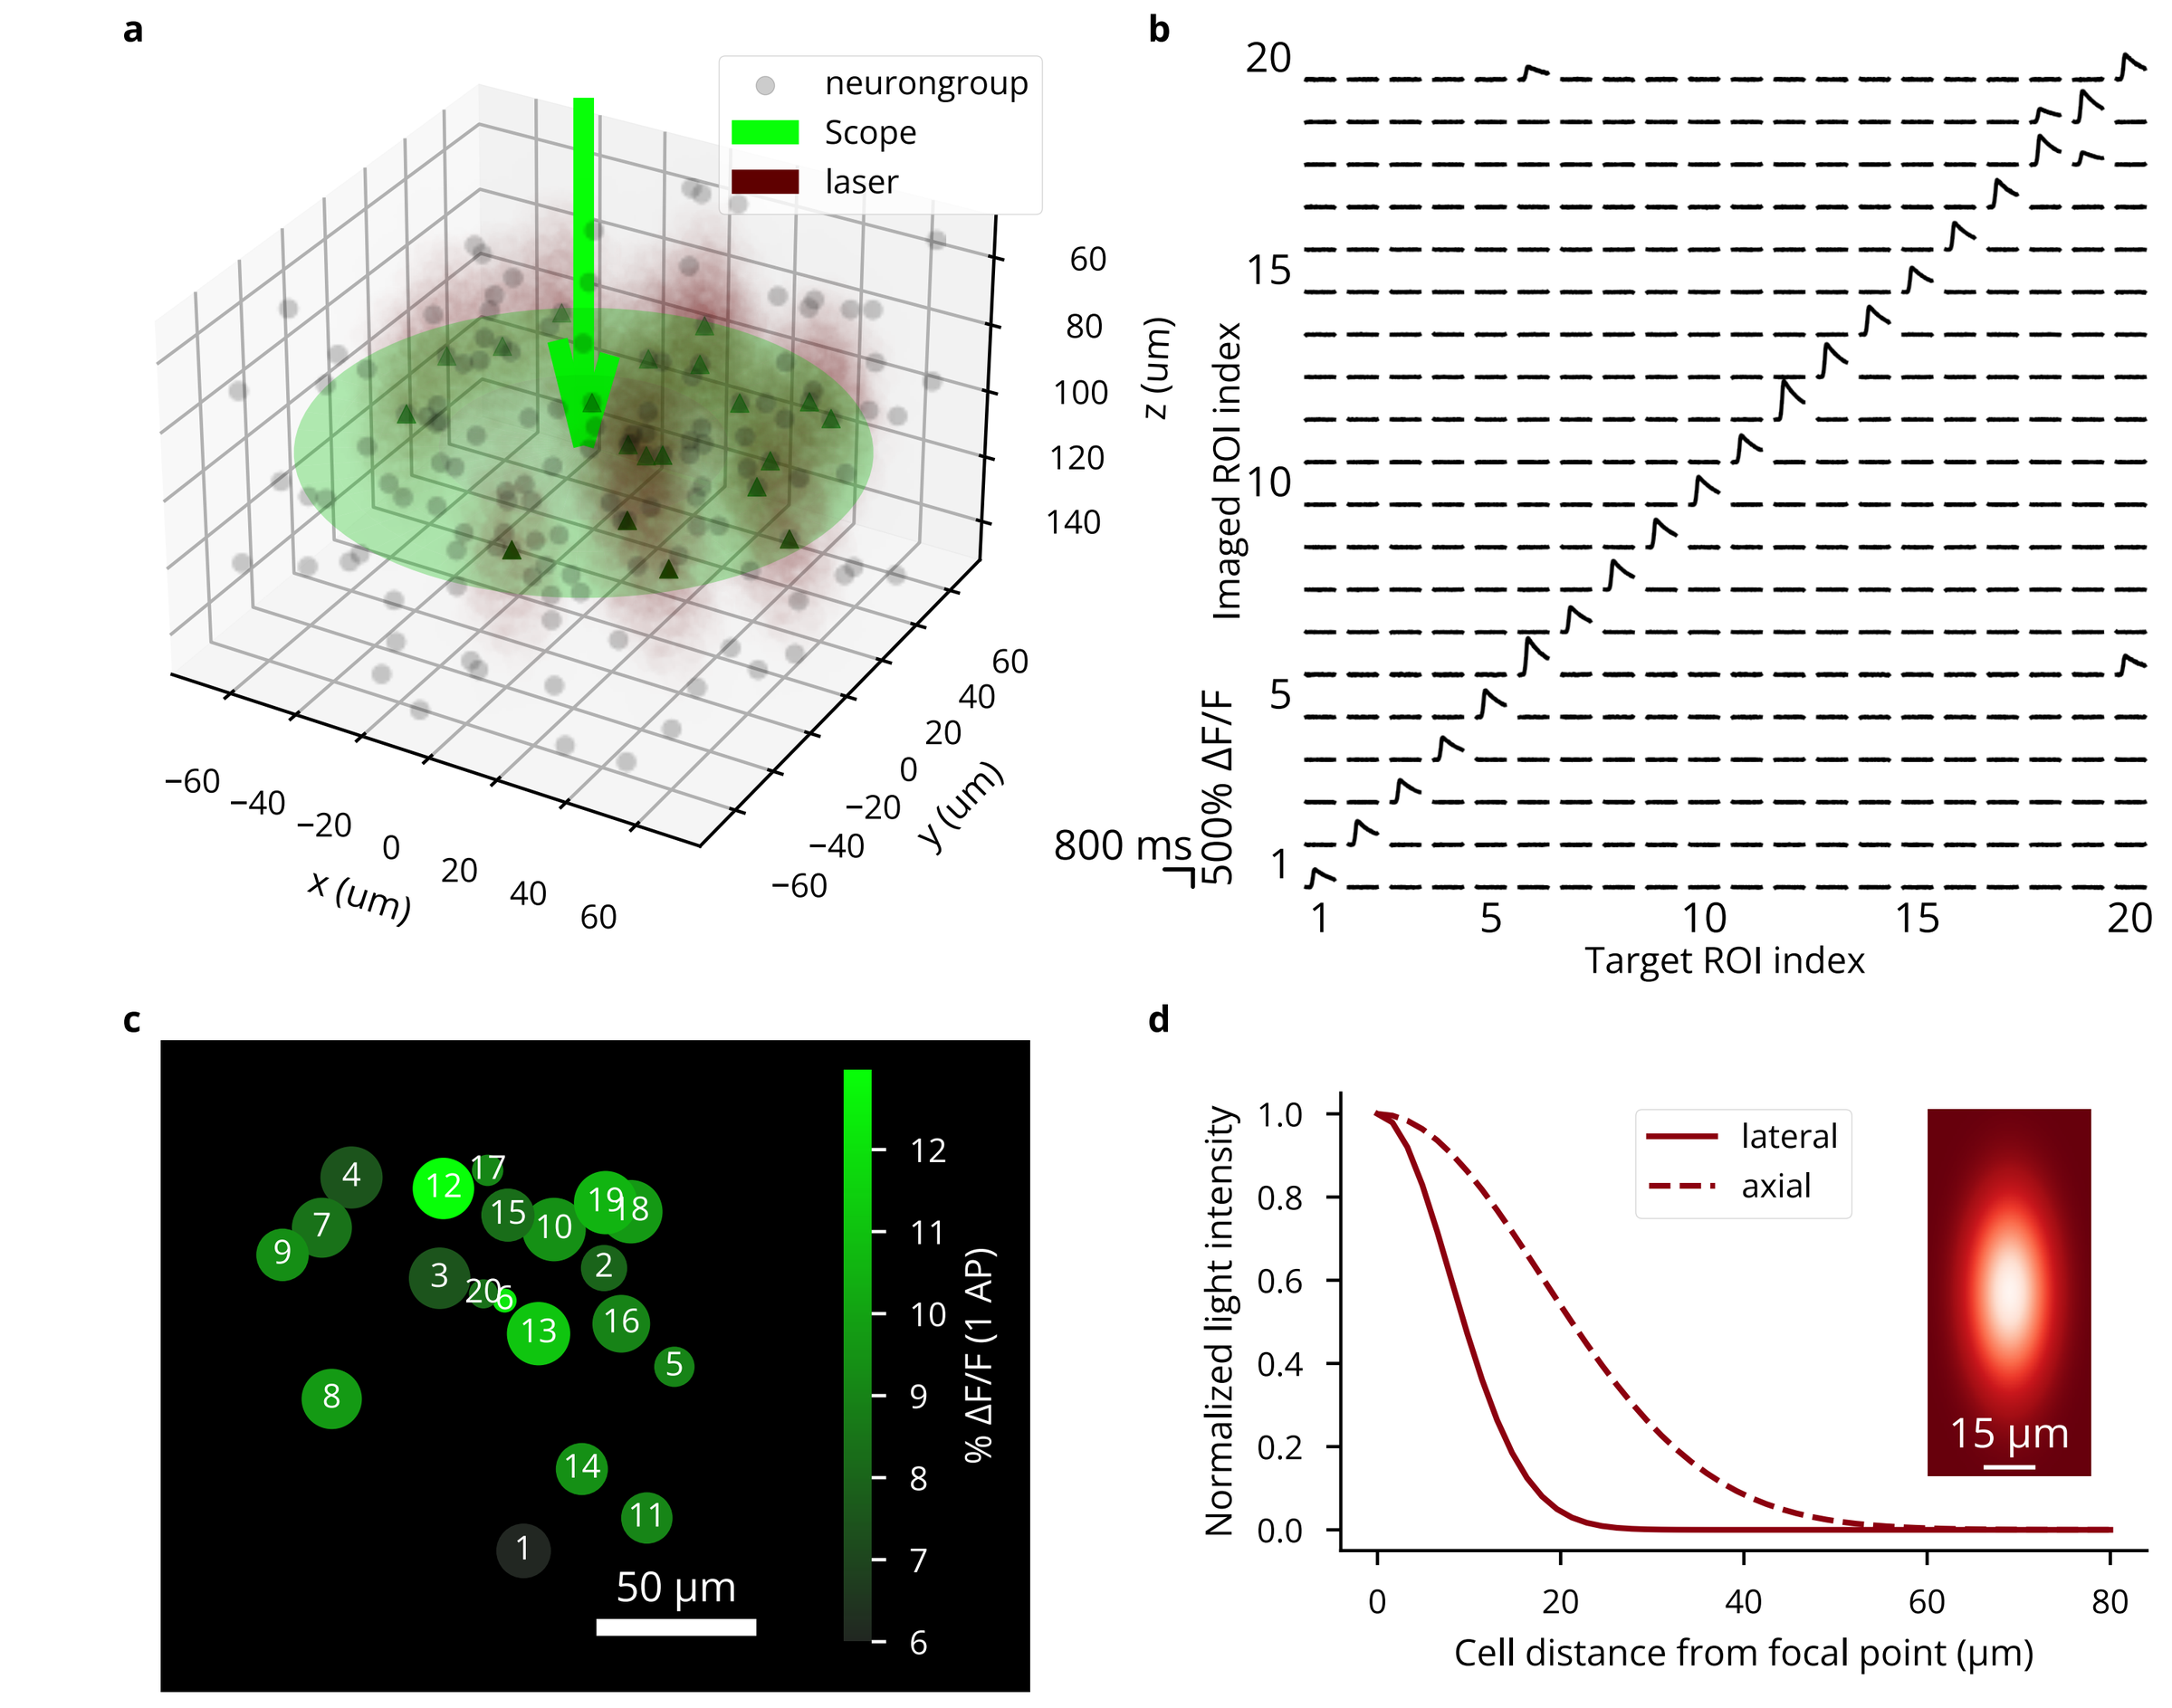
a
neurongroup
Scope
laser
x (um)
y (um)
z (um)
b
Imaged ROI index
Target ROI index
800 ms
500% ΔF/F
c
50 μm
% ΔF/F (1 AP)
d
lateral
axial
Normalized light intensity
Cell distance from focal point (μm)
15 μm

Signal and noise amplitude depend on the indicator, but are scaled by per-ROI expression level and pixels in focus:

$$\mathrm{SNR}_i = \frac{\Delta F/F_{0\mathrm{1AP}}}{\sigma_{\mathrm{noise}}} \frac{\rho_{\mathrm{rel}_i}}{1/\sqrt{N_i}}.$$

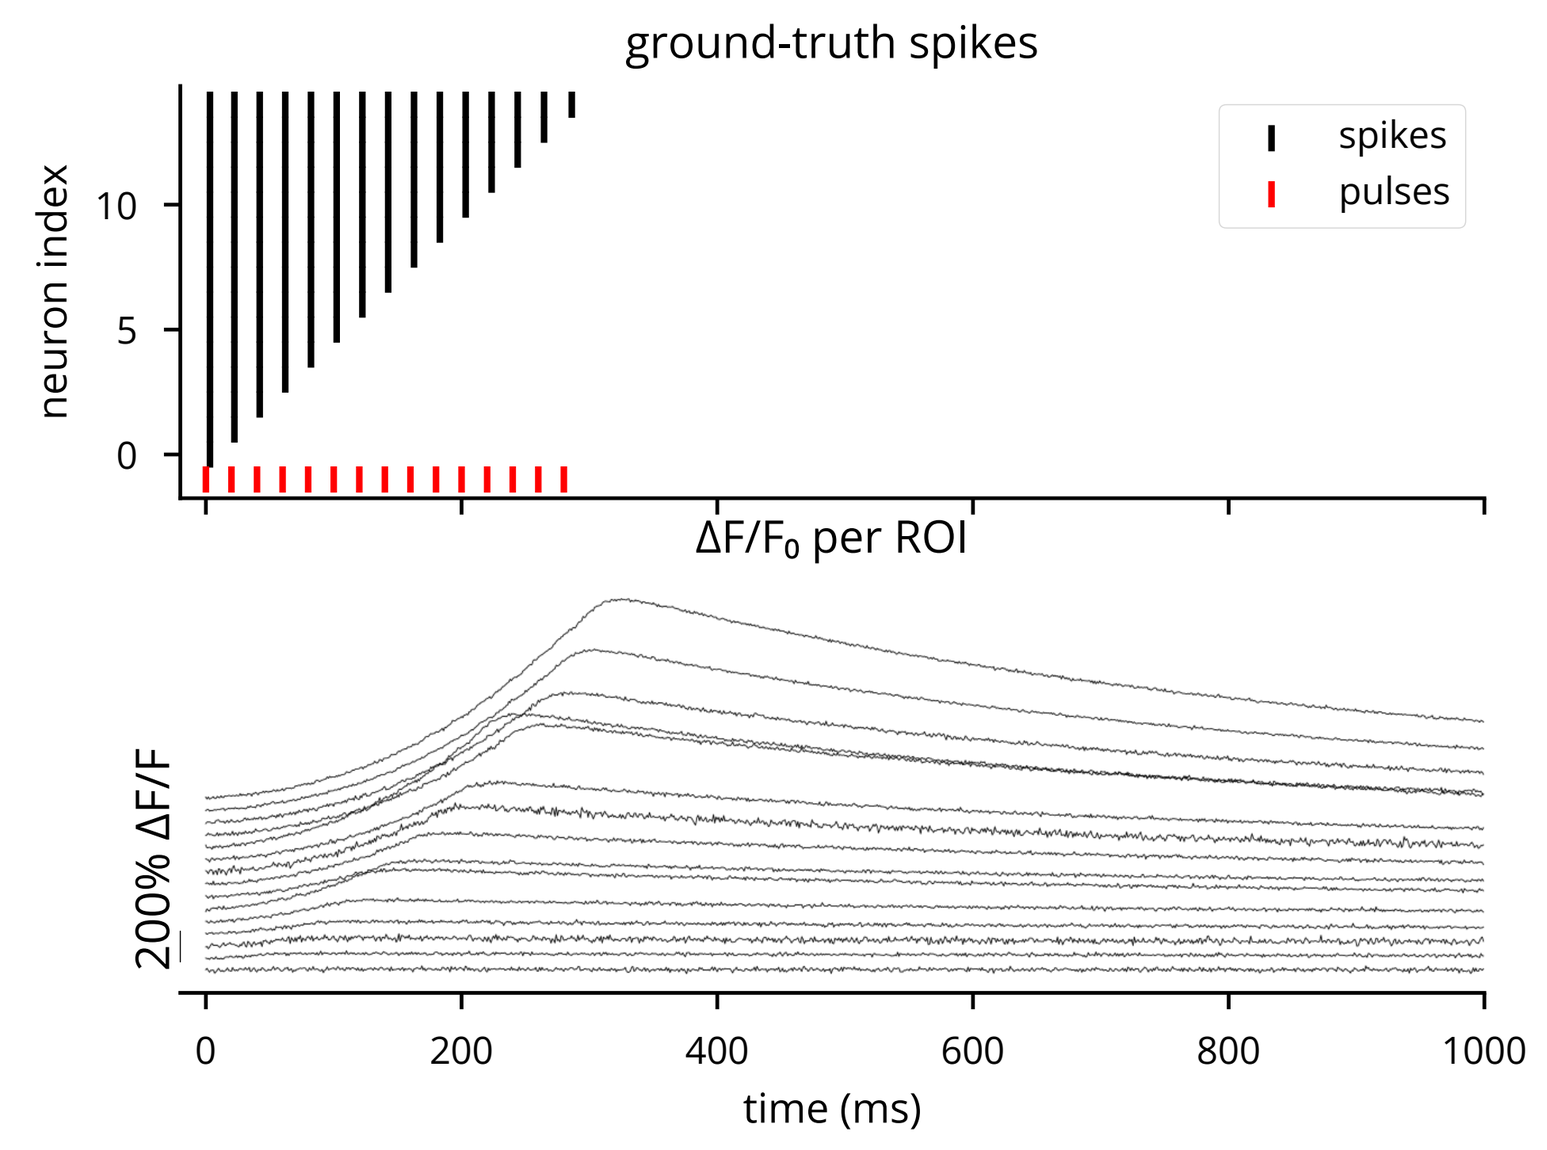
ground-truth spikes
spikes
pulses
neuron index
ΔF/F₀ per ROI
200% ΔF/F
time (ms)

Detailed models of spike-driven calcium and GECI dynamics underlie fluorescence measurements [9]:

$$\frac{\mathrm{d}\left[\mathrm{Ca}^{2+}\right]}{\mathrm{d}t} = -\gamma \frac{\left[\mathrm{Ca}^{2+}\right] - \left[\mathrm{Ca}^{2+}\right]_{\mathrm{rest}}}{1+\kappa_S+\kappa_B} \qquad \Delta\left[\mathrm{Ca}^{2+}\right]\left(t_{\mathrm{spike}}\right) = \frac{\Delta\left[\mathrm{Ca}^{2+}\right]_T}{1+\kappa_S+\kappa_B}$$

Interoperable data export via Neo

Neo export of simulation data facilitates downstream analysis [10]:

```
>>> sim.to_neo()
Block with 1 segments, 1 groups
description: 'Exported from Cleo simulation'
annotations: {'export_datetime': datetime.datetime(2023, 9, 6, 9, 59, 33, 320150)}
# segments (N=1)
0: Segment with 2 analogsignals, 58 spiketrains
   # analogsignals (N=2)
   0: AnalogSignal with 2 channels of length 50; units mW/mm**2; datatype float64
      name: 'Light'
      description: 'Exported from Cleo Light device'
      annotations: {'export_datetime': datetime.datetime(2023, 9, 6, 9, 59, 33, 320674)}
      sampling rate: 1.0 1/ms
      time: 0.0 ms to 50.0 ms
   1: AnalogSignal with 3 channels of length 50; units uV; datatype float64
      name: 'Probe.TKLFPSignal'
      description: 'Exported from Cleo TKLFPSignal object'
      annotations: {'export_datetime': datetime.datetime(2023, 9, 6, 9, 59, 33, 321221)}
      sampling rate: 1.0 1/ms
      time: 0.0 ms to 50.0 ms
```

Case studies

Optogenetically clamping PV firing rate disrupts plasticity in a V1 model [11]:

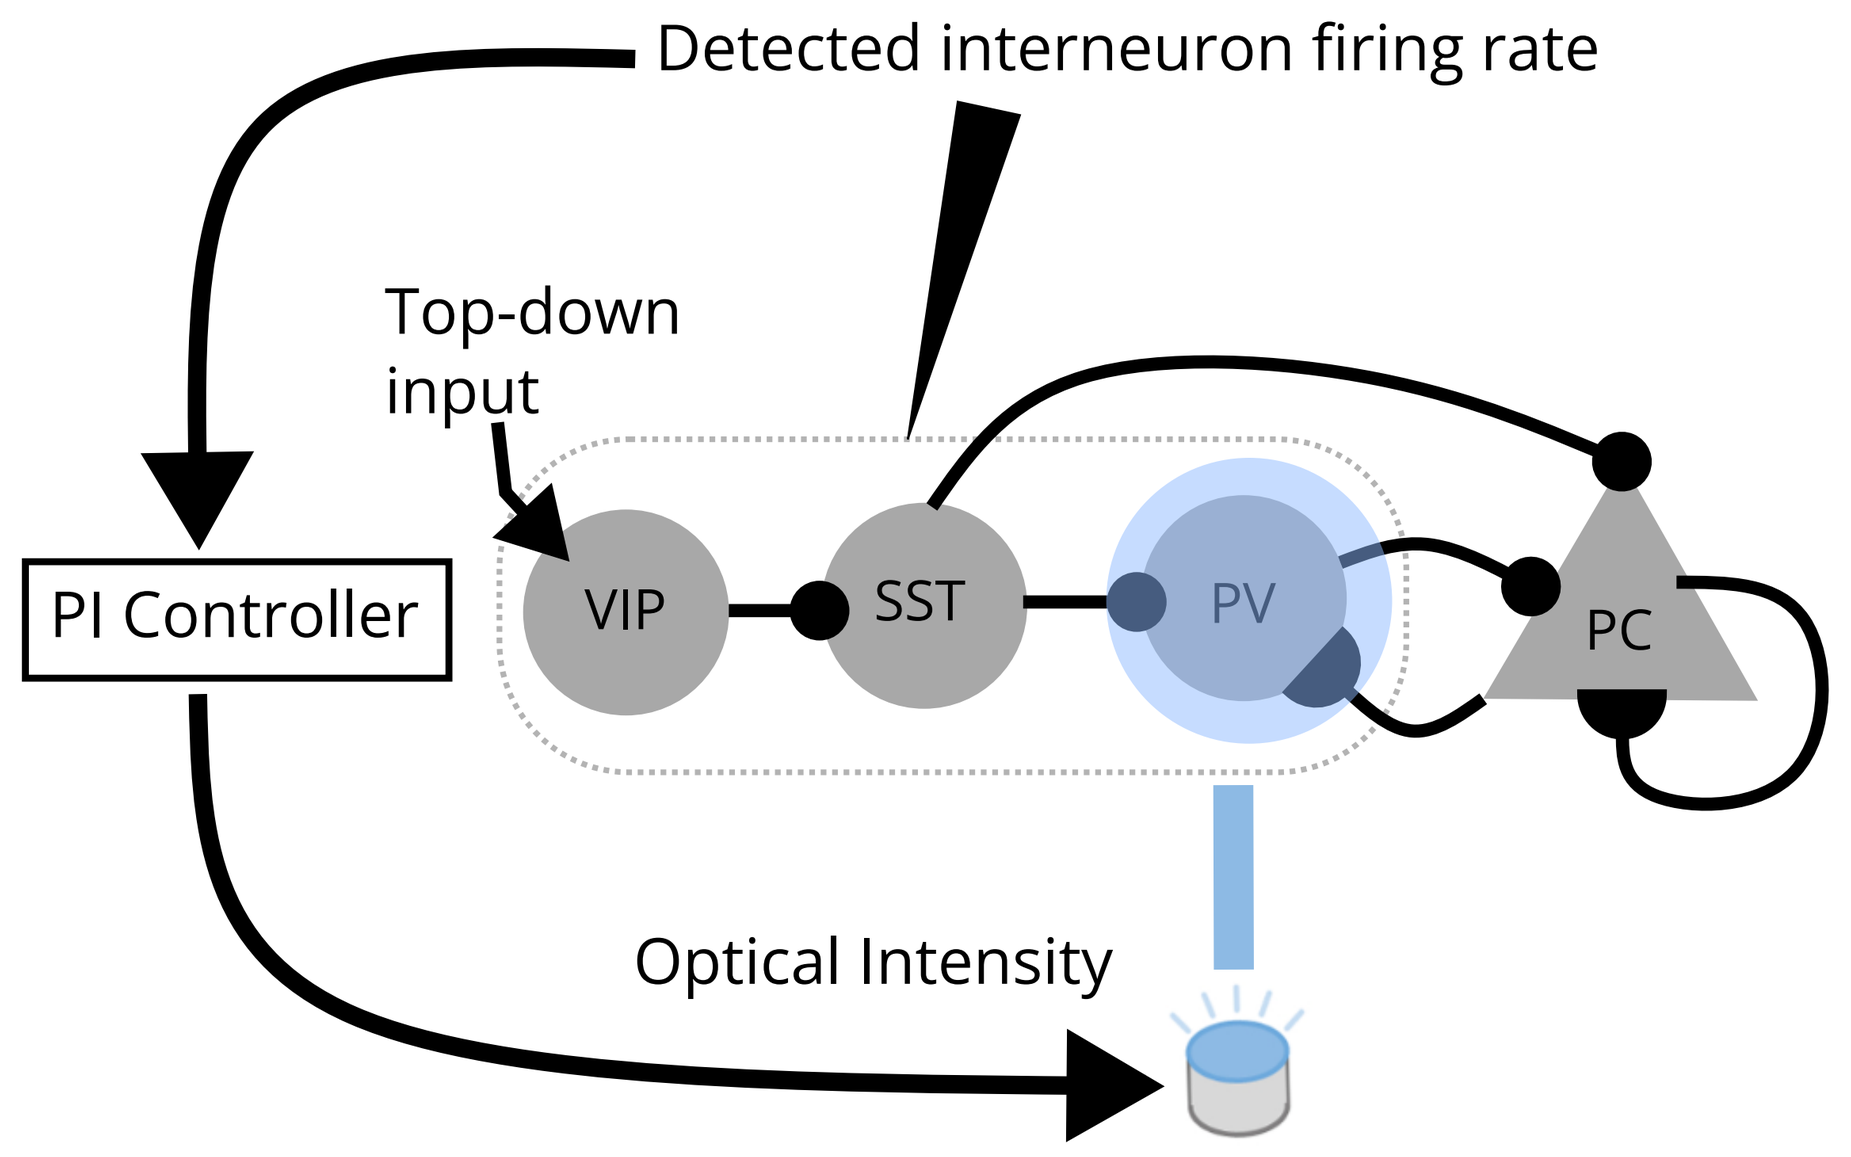
Detected interneuron firing rate
Top-down input
PI Controller
VIP
SST
PV
PC
Optical Intensity

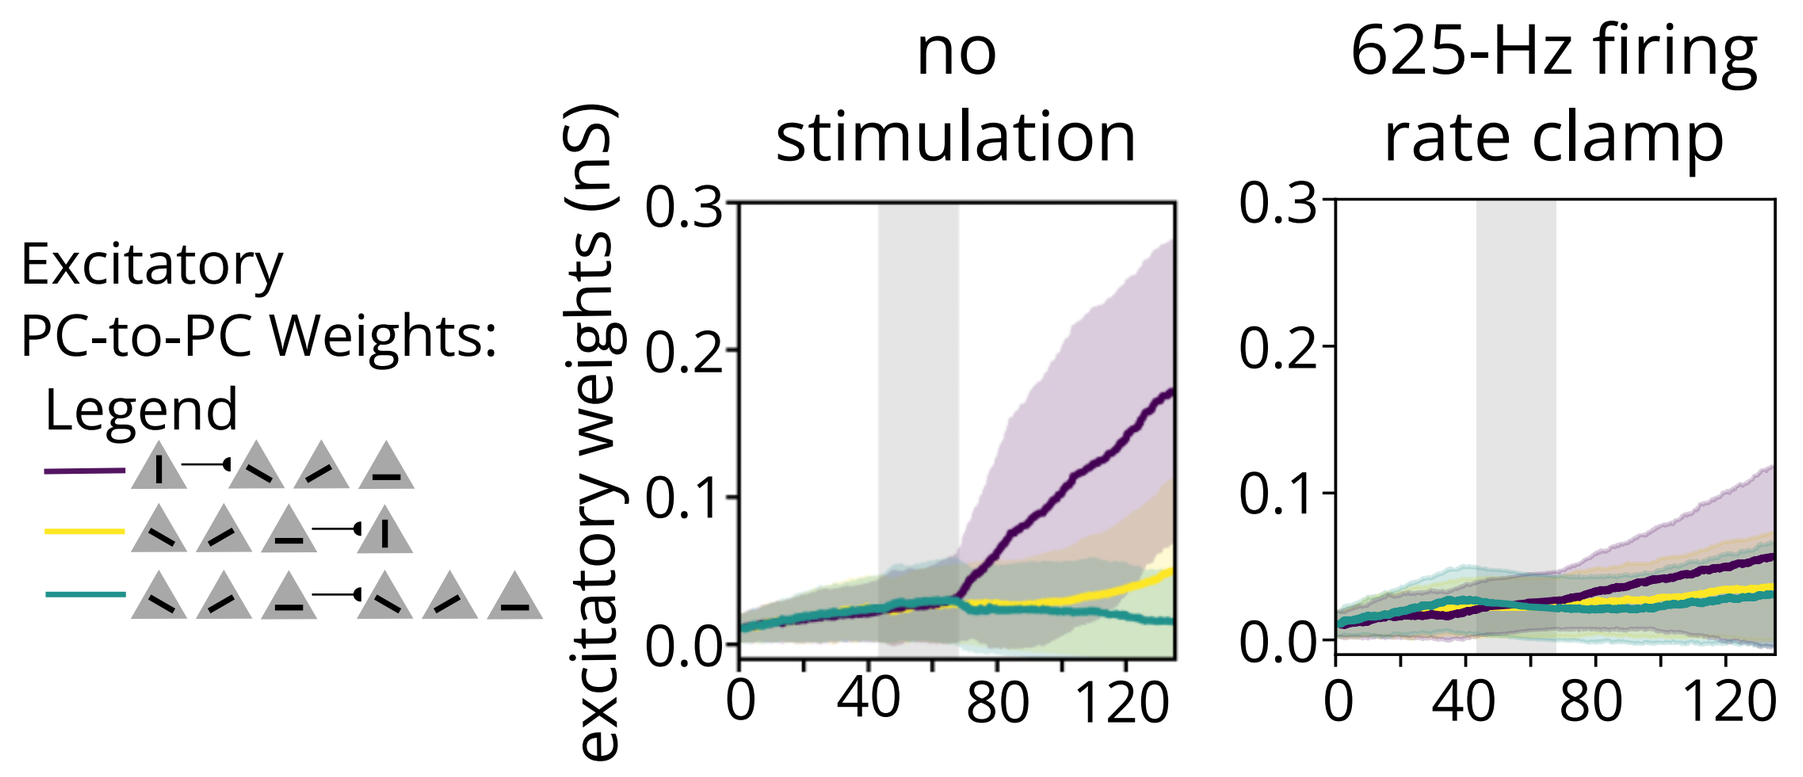
Excitatory PC-to-PC Weights: Legend
no stimulation
625-Hz firing rate clamp
excitatory weights (nS)

CLOC can evoke a reference SWR-like oscillation in a HPC model [12] without manual stimulus design or calibration:

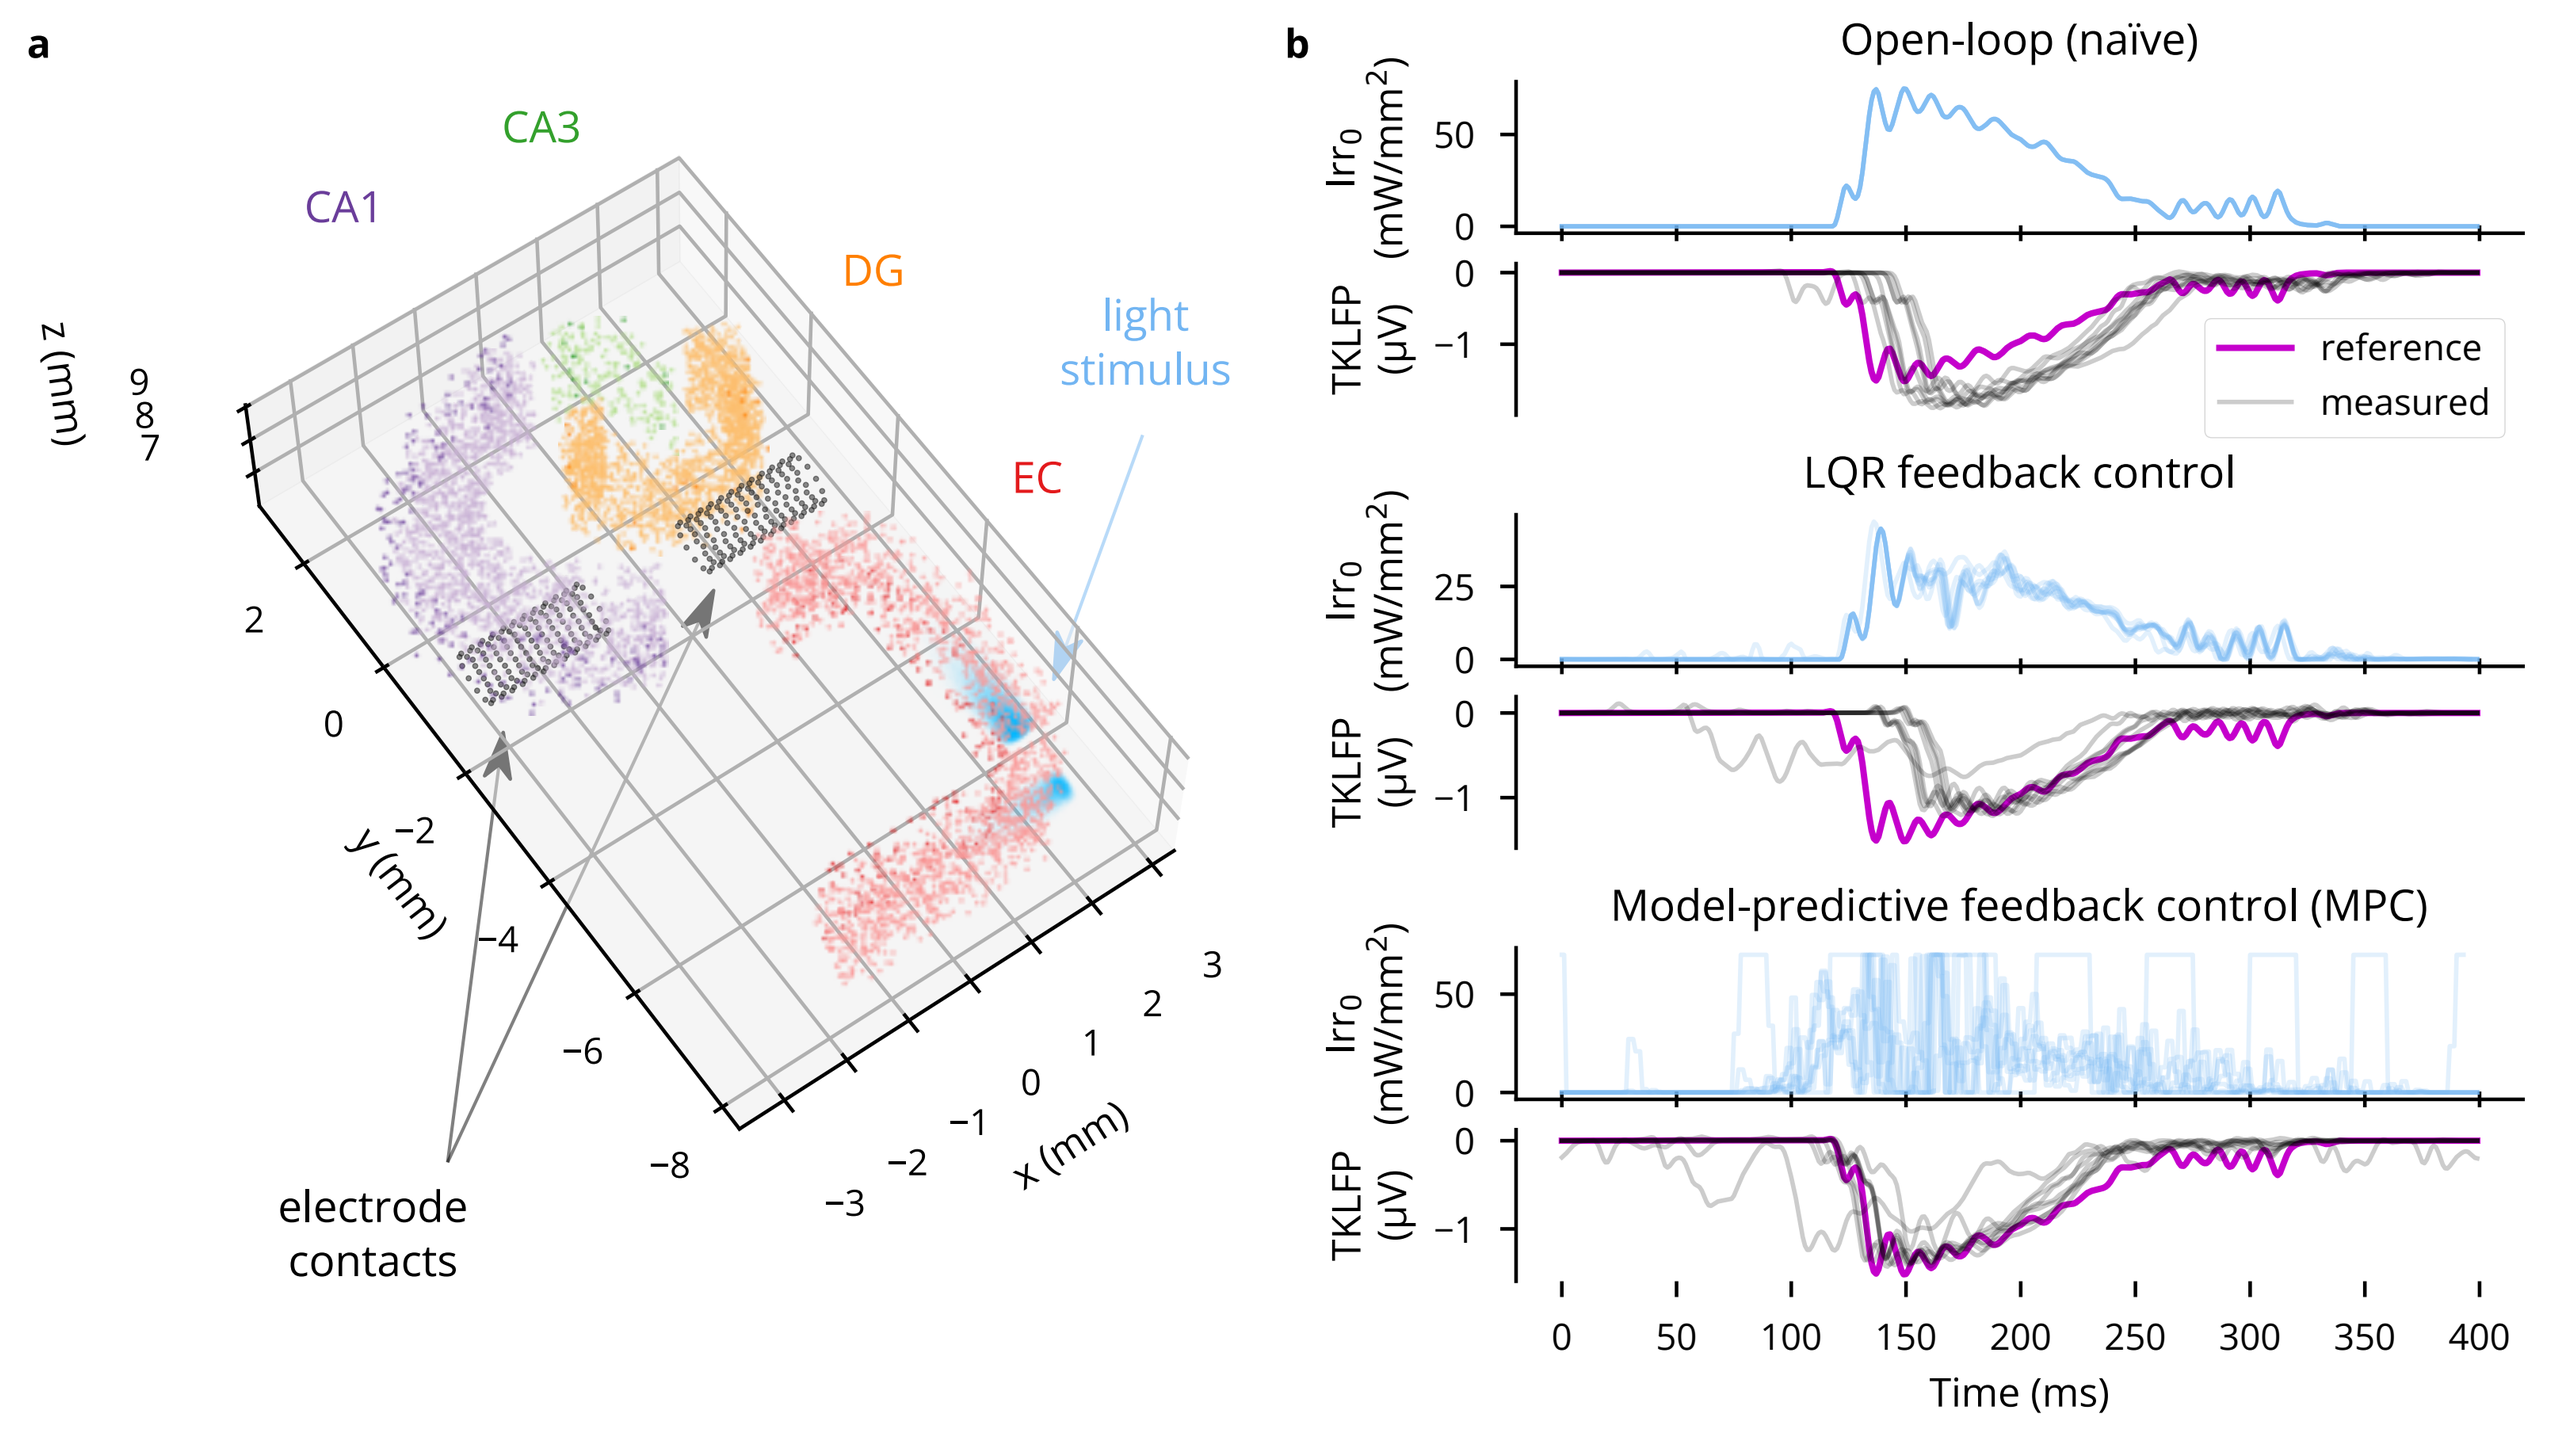
a
CA1
CA3
DG
EC
light stimulus
electrode contacts
x (mm)
y (mm)
z (mm)
b
Open-loop (naïve)
reference
measured
LQR feedback control
Model-predictive feedback control (MPC)
Time (ms)

Acknowledgments

Supported by NIH CRCNS grant R01NS115327.
Kyle Johnsen was supported by the Georgia Tech/Emory NIH/NIBIB Training Program in Computational Neural Engineering (T32EB025816).

References

[1] A A Willats, M R O'Shaughnessy, and C J Rozell. Closed-loop identifiability in neural circuits. *In prep*, 2023.
[2] A A Willats, M F Bolus, K A Johnsen, G B Stanley, and C J Rozell. Cloc tools: A library of tools for closed-loop neuroscience. *In prep*, 2023.
[3] A A Willats, M F Bolus, C J Whitmire, G B Stanley, and C J Rozell. State-aware control of switching neural dynamics. *In prep*, 2023.
[4] M F Bolus, A A Willats, C J Whitmire, C J Rozell, and G B Stanley. Design strategies for dynamic closed-loop optogenetic neurocontrol in vivo. *Journal of Neural Engineering*, 15(2):026011, April 2018. ISSN 1741-2560. doi: 10.1088/1741-2552/aaa506.
[5] M F Bolus, A A Willats, C J Rozell, and G B Stanley. State-space optimal feedback control of optogenetically driven neural activity. *Journal of neural engineering*, 18(3): 036006, June 2021. doi: 10.1101/2020.06.25.171785.
[6] Bartosz Telenczuk, Maria Telenczuk, and Alain Destexhe. A kernel-based method to calculate local field potentials from networks of spiking neurons. *Journal of Neuroscience Methods*, 344:108871, October 2020. ISSN 1872678X. doi: 10.1016/j.jneumeth.2020.108871.
[7] Thomas J Foutz, Richard L Arlow, and Cameron C Mcintyre. Theoretical principles underlying optical stimulation of a channelrhodopsin-2 positive pyramidal neuron. *J Neurophysiol*, 107:3235–3245, 2012. doi: 10.1152/jn.00501.2011.-Optogenetics.
[8] Himanshu Bansal, Neha Gupta, and Sukhdev Roy. Theoretical Analysis of Low-power Bidirectional Optogenetic Control of High-frequency Neural Codes with Single Spike Resolution. *Neuroscience*, 449:165–188, November 2020. ISSN 18737544. doi: 10.1016/j.neuroscience.2020.09.022.
[9] Alexander Song, Jeff L. Gauthier, Jonathan W. Pillow, David W. Tank, and Adam S. Charles. Neural anatomy and optical microscopy (NAOMi) simulation for evaluating calcium imaging methods. *Journal of Neuroscience Methods*, 358:109173, July 2021. ISSN 0165-0270. doi: 10.1016/j.jneumeth.2021.109173.
[10] Samuel Garcia, Domenico Guarino, Florent Jaillet, Todd Jennings, Robert Pröpper, Philipp L. Rautenberg, Chris C. Rodgers, Andrey Sobolev, Thomas Wachtler, and Pierre Yger. Neo: An object model for handling electrophysiology data in multiple formats. *Frontiers in neuroinformatics*, 8:10, 2014.
[11] Katharina Anna Wilmes and Claudia Clopath. Inhibitory microcircuits for top-down plasticity of sensory representations. *Nature Communications*, 10(1):5055, November 2019. ISSN 2041-1723. doi: 10.1038/s41467-019-12972-2.
[12] Amélie Aussel, Laure Buhry, Louise Tyvaert, and Radu Ranta. A detailed anatomical and mathematical model of the hippocampal formation for the generation of sharp-wave ripples and theta-nested gamma oscillations. *Journal of Computational Neuroscience*, 45(3):207–221, December 2018. ISSN 1573-6873. doi: 10.1007/s10827-018-0704-x.